Injectable cross-linked rhCollagen scaffold combined with PRP for soft tissue repair

Racheli Gueta, PhD(1), Frida Grynspan, PhD(2), Tal Amzel, MSc(1), Jasmine Seror, PhD(1), Ofer Levy, MD MCh(Orth) FRCS(3), Nadav Orr, PhD(1) ,Oded Shoseyov, PhD(1,4)

[1]CollPlant Ltd., Ness-Ziona, Israel;
[2]Stem Cell Medicine Ltd, Jerusalem, Israel;
[3]Reading Shoulder Unit, the Royal Berkshire Hospital & the Berkshire Independent Hospital, Reading, UK. The Centre for Sports Medicine and Human Performance, School of Sport and Education Brunel University, London, UK;
[4]The Robert H. Smith Faculty of Agricultural, Food and Environmental Quality Sciences, The Hebrew University of Jerusalem, Rehovot, Israel.

Introduction

CollPlant has generated a bio-engineered plant line capable of expressing human recombinant type I collagen.

Vergenix™STR is an injectable gel composed of cross-linked human recombinant type I collagen used in combination with autologous platelet rich plasma (PRP).

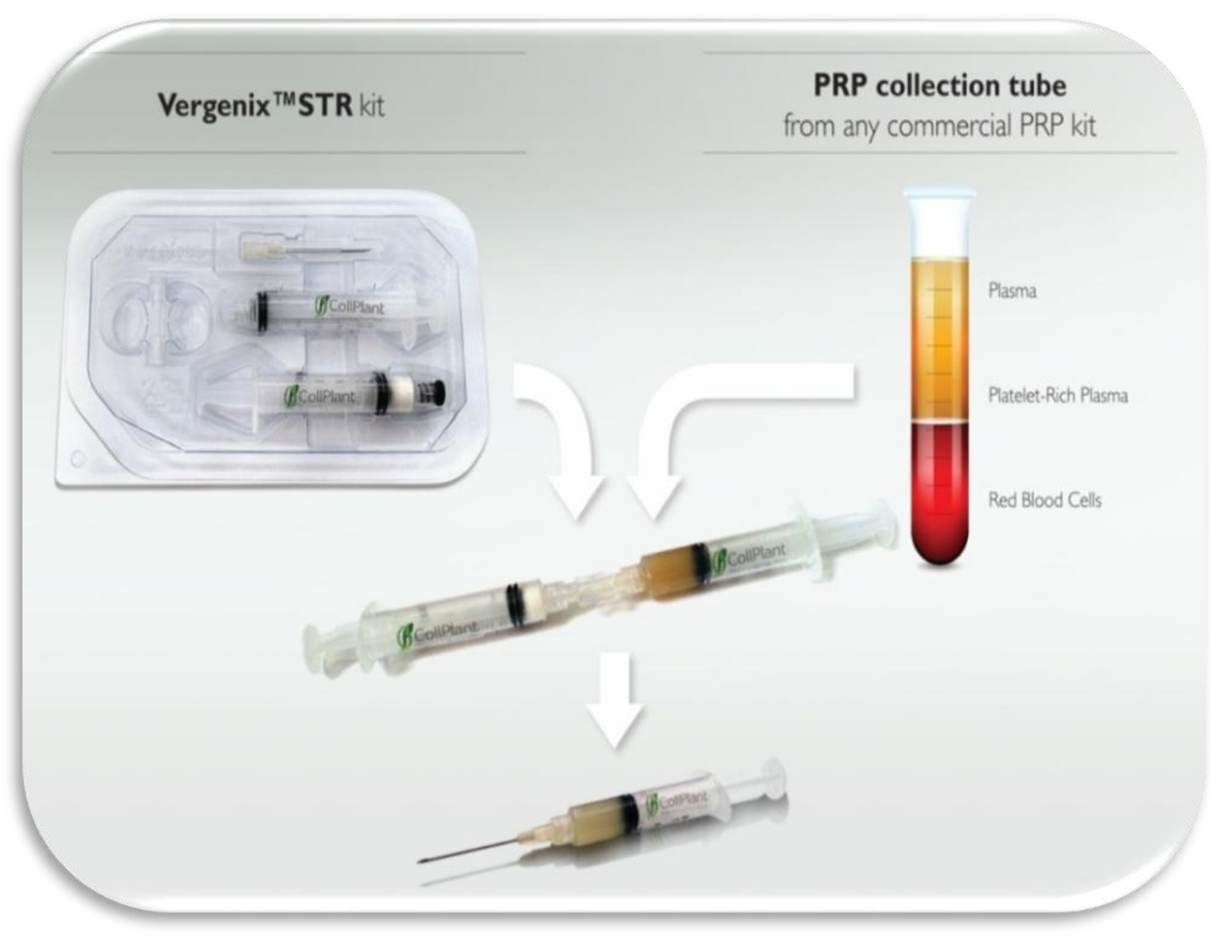

Vergenix™STR kit
PRP collection tube
from any commercial PRP kit

Upon injection, the complex forms a collagen-fibrin matrix at the injury site and promotes cell migration and tissue repair.

Vergenix™ STR combined with PRP (STR/PRP) is now in clinical trial for the treatment of tendinopathy.

In vitro study: cell proliferation

Normal human dermal fibroblasts (nHDF) were seeded on STR/PRP matrix and on thrombin activated PRP under starvation conditions.

- ✓ Cell viability is significantly higher on STR/PRP matrix than on PRP clot or starvation medium (control)
- ✓ STR/PRP promotes cell proliferation

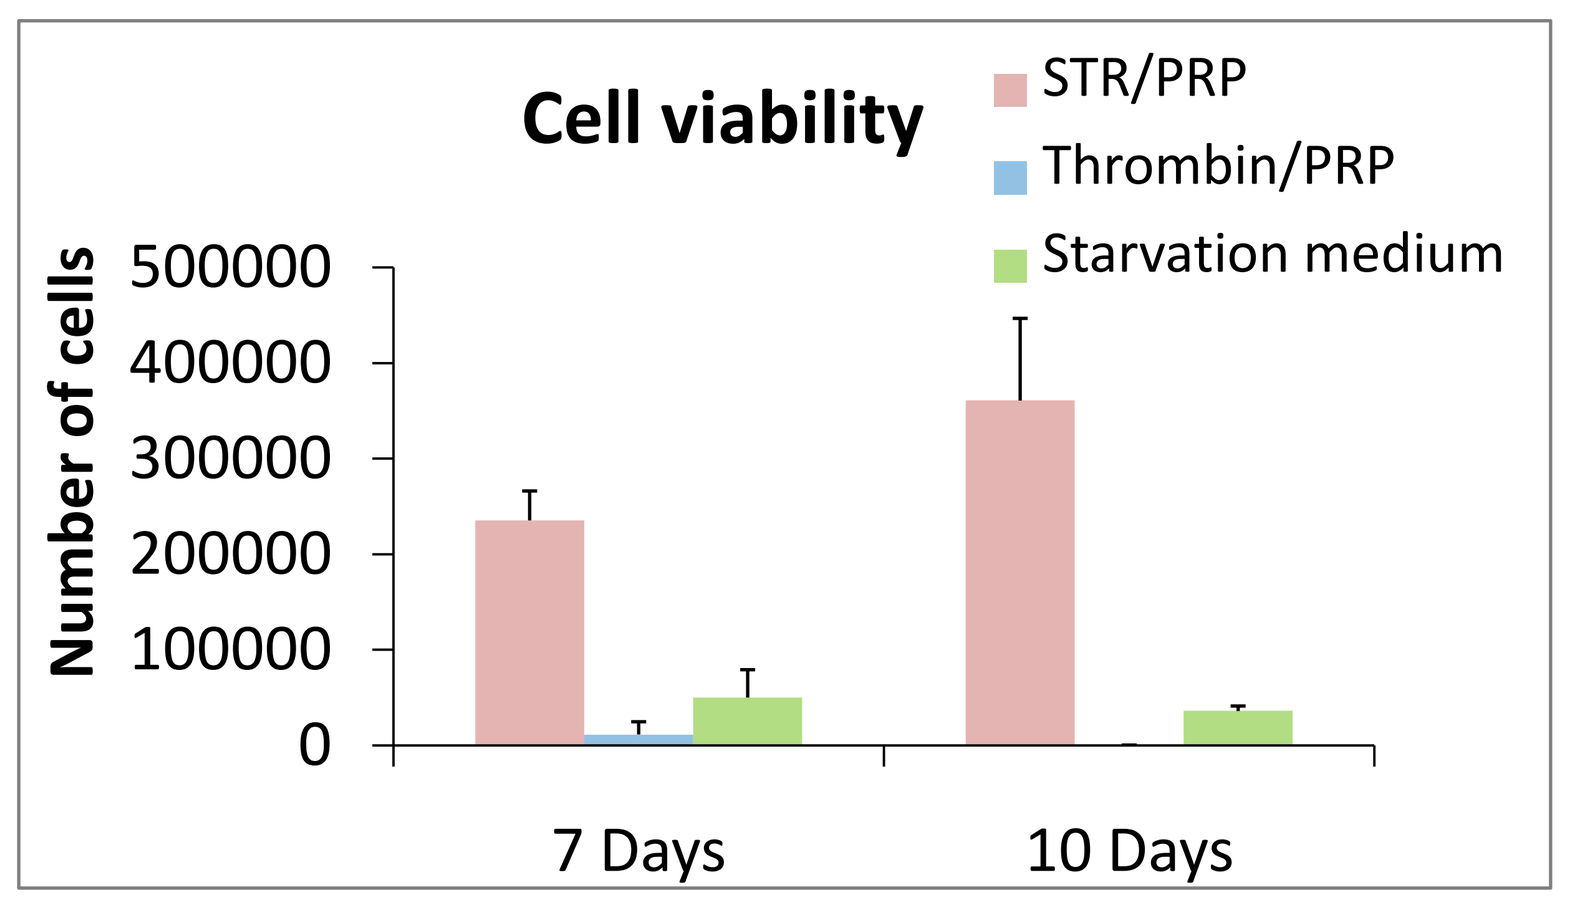

Cell viability
STR/PRP
Thrombin/PRP
Starvation medium
Number of cells
500000
400000
300000
200000
100000
0
7 Days
10 Days

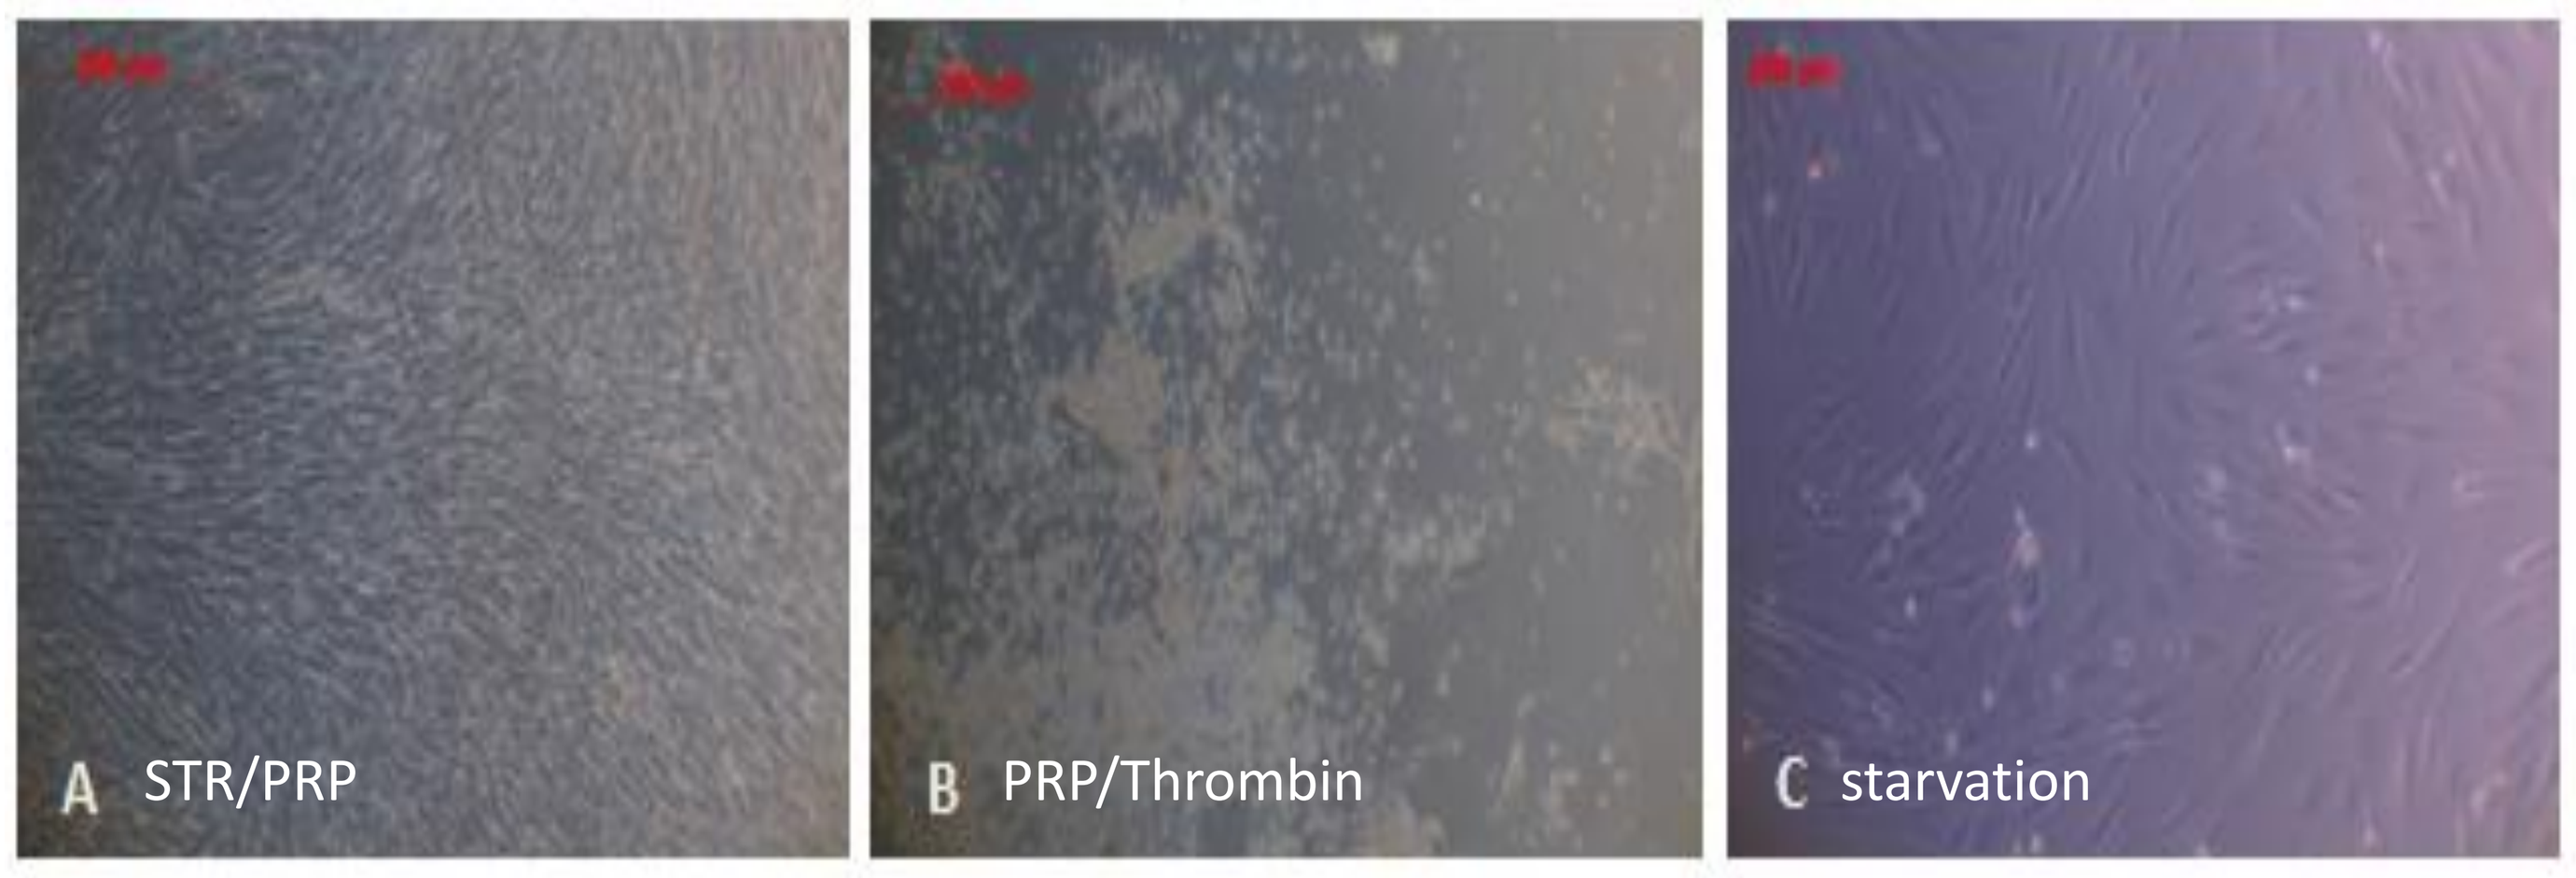

A STR/PRP
B PRP/Thrombin
C starvation

In vivo study: matrix degradation and growth factors (GFs) release in a subcutaneous rat model

Vergenix™ STR combined with PRP and activated PRP alone were injected subcutaneously in rats. Matrix degradation (weight) and GFs release (GFs content measured by ELISA) were assessed at several time points (0, 7, 14, 21, 30, 45 days)

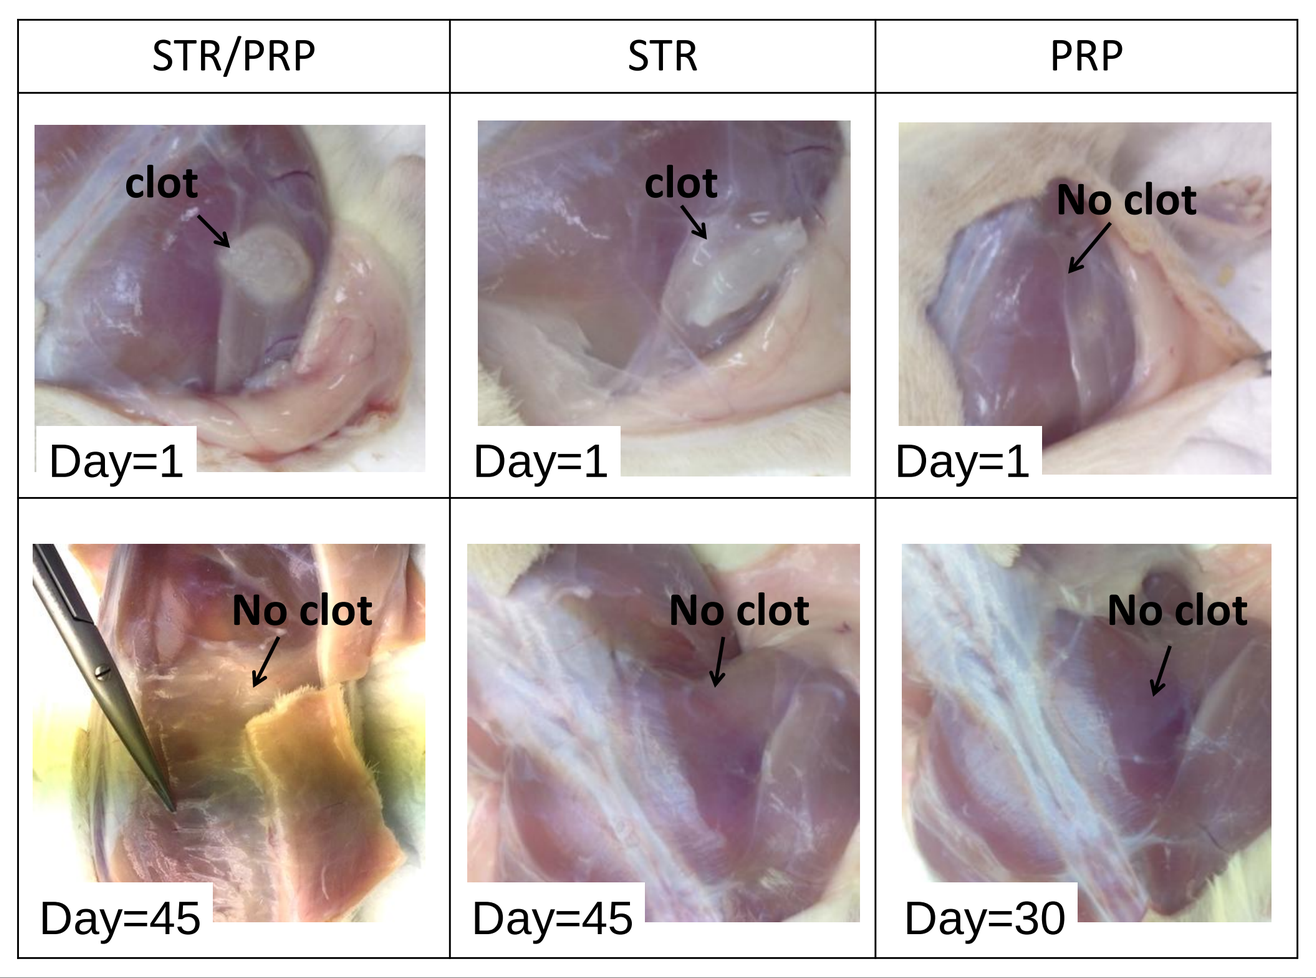

STR/PRP
STR
PRP
clot
clot
No clot
Day=1
Day=1
Day=1
No clot
No clot
No clot
Day=45
Day=45
Day=30

Degradation

- ✓ STR/PRP gradually degrades over time
- ✓ STR/PRP weight at d=45 is <0.2% of its original weight
- ✓ PRP alone disappears already at day 1

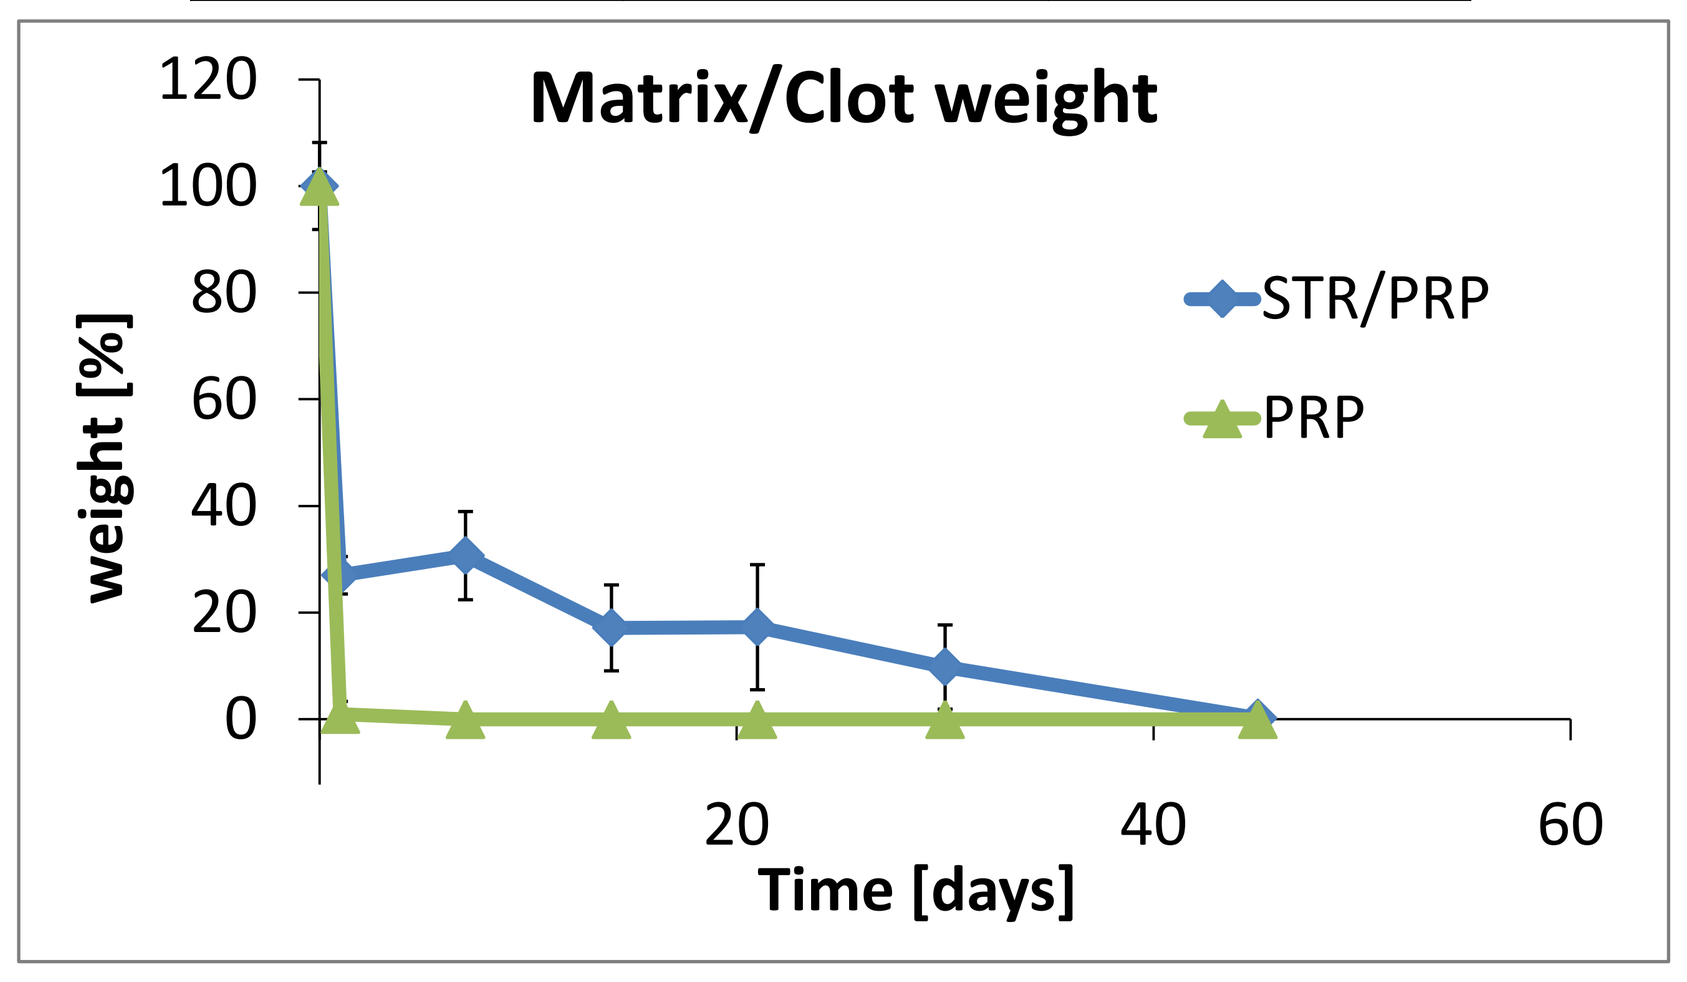

Matrix/Clot weight
weight [%]
Time [days]
STR/PRP
PRP

GFs release: GFs content over time

- ✓ PRP clot: GFs are released in the first 24 hours
- ✓ STR/PRP matrix: the GFs content profile shows a first burst between day 0 and 1 followed by an increase up to day 21 and a consequent decrease up to day 45
- ✓ STR/PRP promotes temporary migration of cells which express high level of platelet-derived growth factors (PDGFs) and vascular endothelial growth factors (VEGFs)

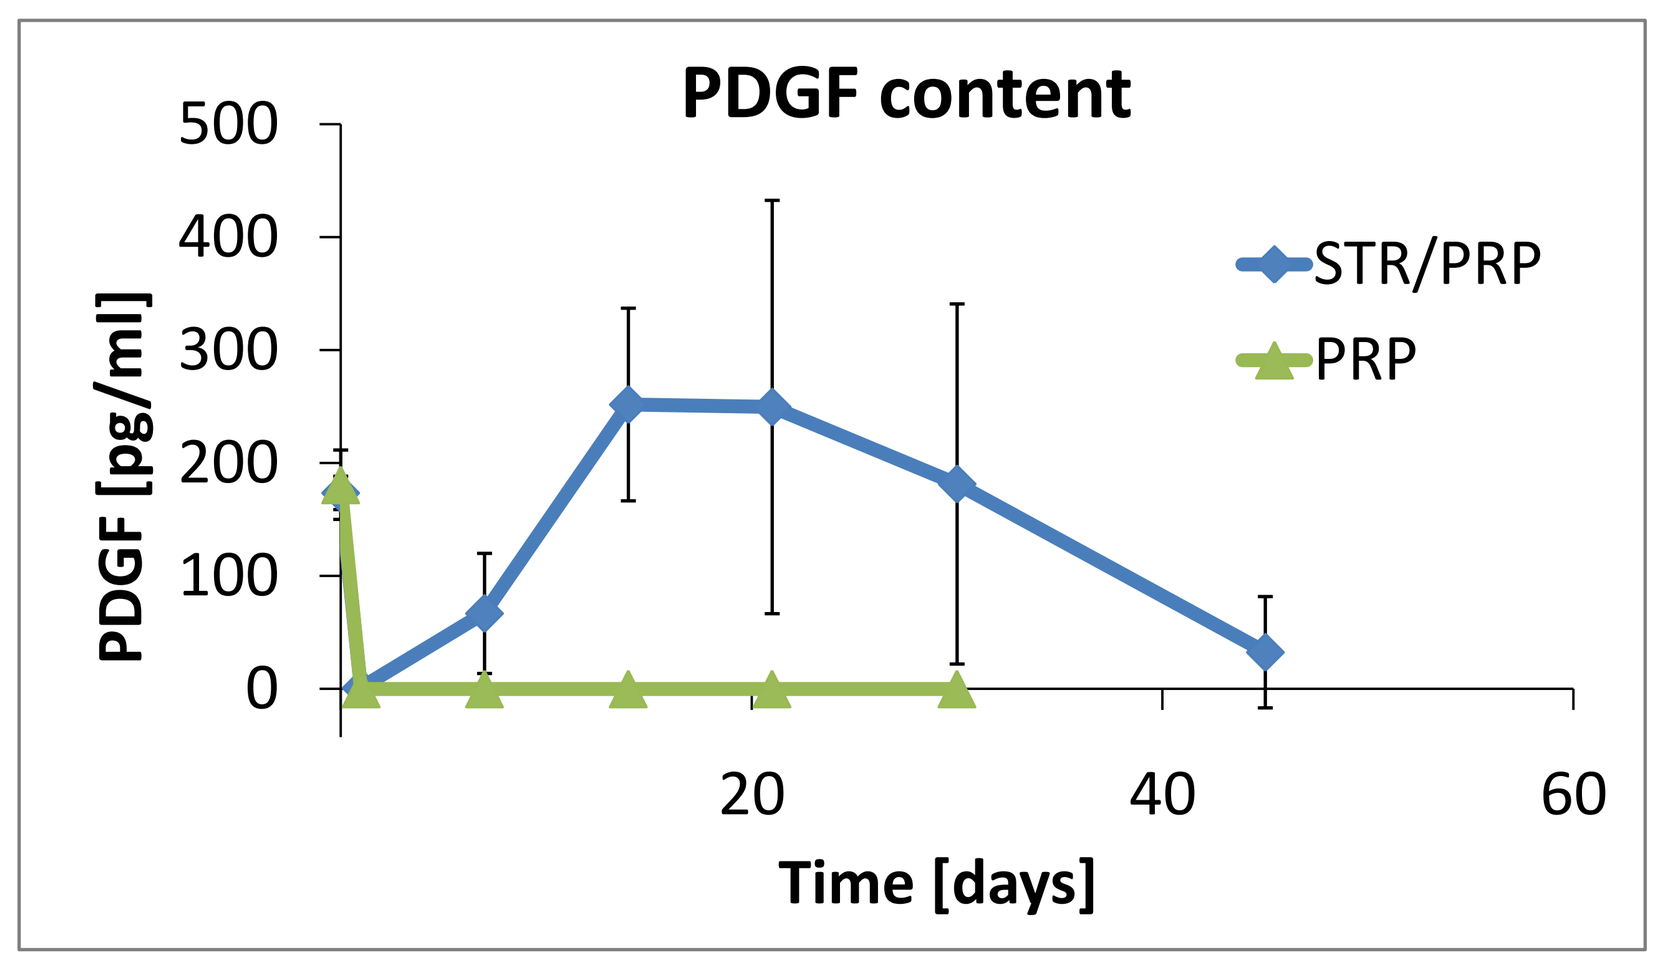

PDGF content
PDGF [pg/ml]
Time [days]
STR/PRP
PRP

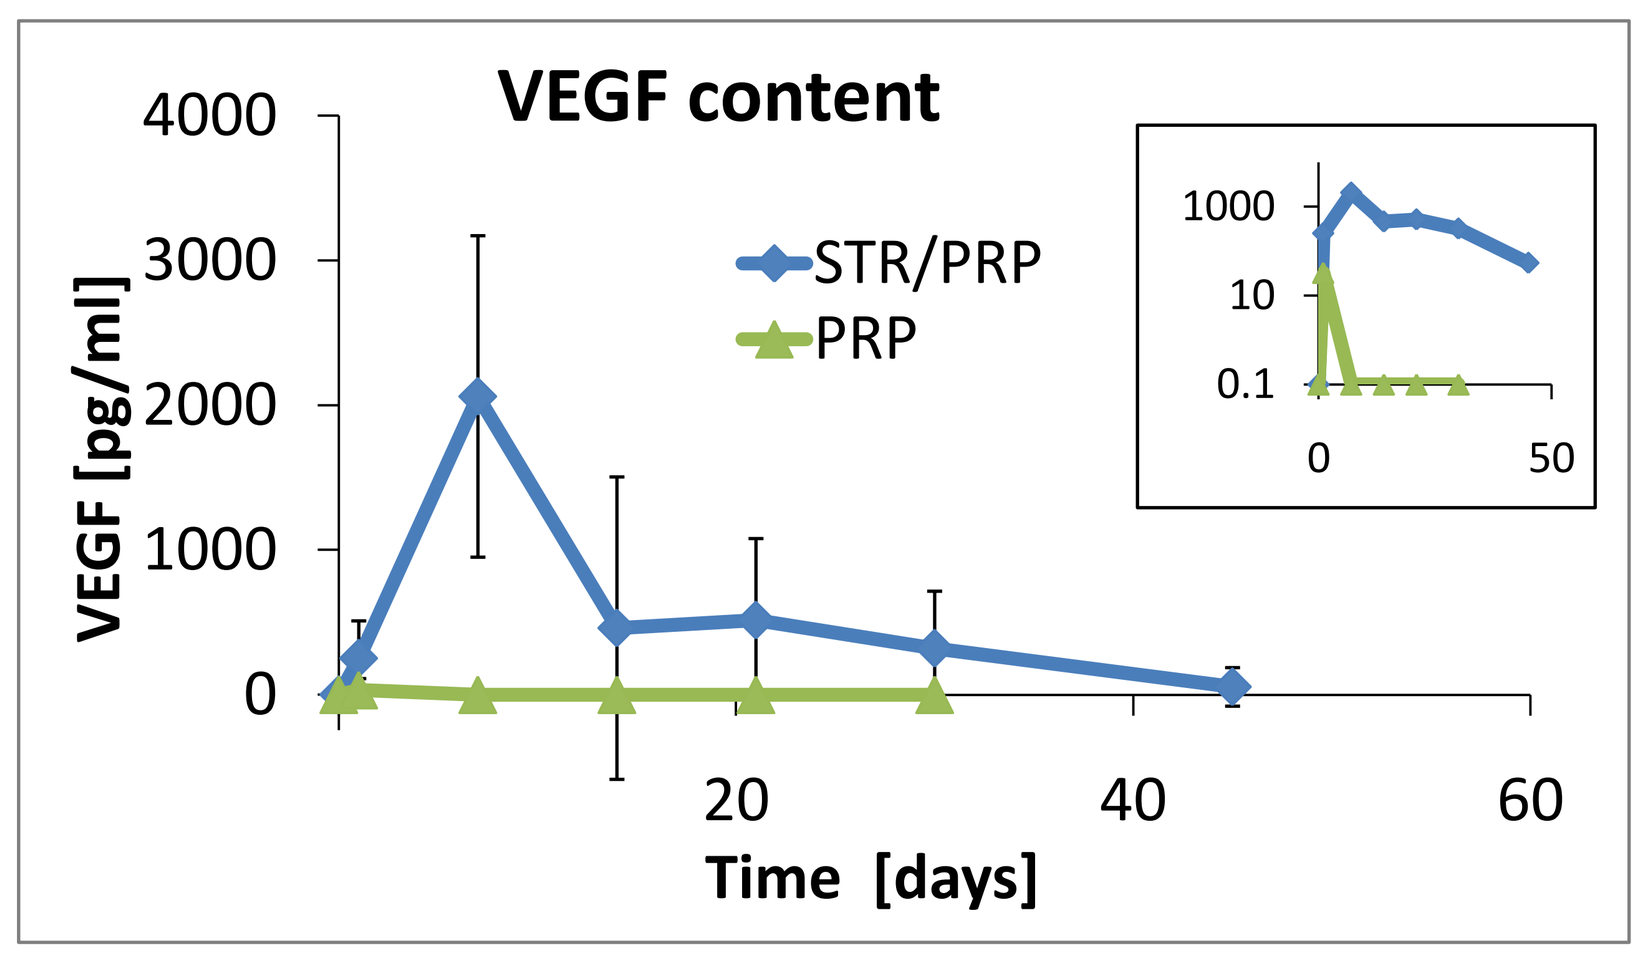

VEGF content
VEGF [pg/ml]
Time [days]
STR/PRP
PRP

In vivo study: Achilles tendon tendinopathy model in rats

Tendinopathy was induced by collagenase injection into the right Common Calcaneal rat's tendon.

Treatment was administered a week later, by injection into the same tendon.

Clinical pathology evaluation and histopathological evaluation were performed after 3, 7 and 14 days.

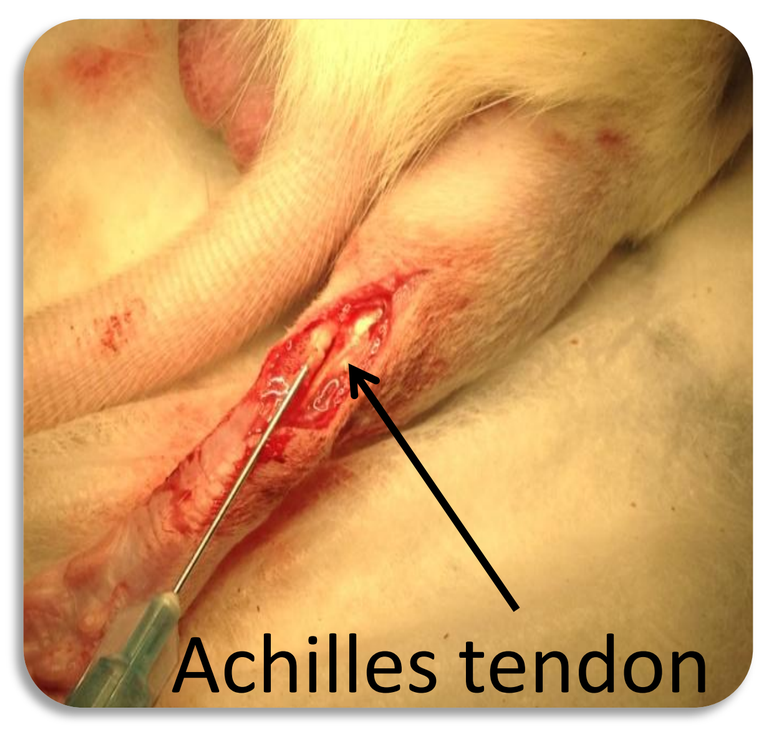

Achilles tendon

Vergenix™ STR combined with PRP promotes:

- ✓ Decrease in the incidence of inflammatory mononuclear cells
- ✓ Reduction in the severity of immature granulation tissue (i.e. fibroplasia)
- ✓ Increase in mature fibrosis.

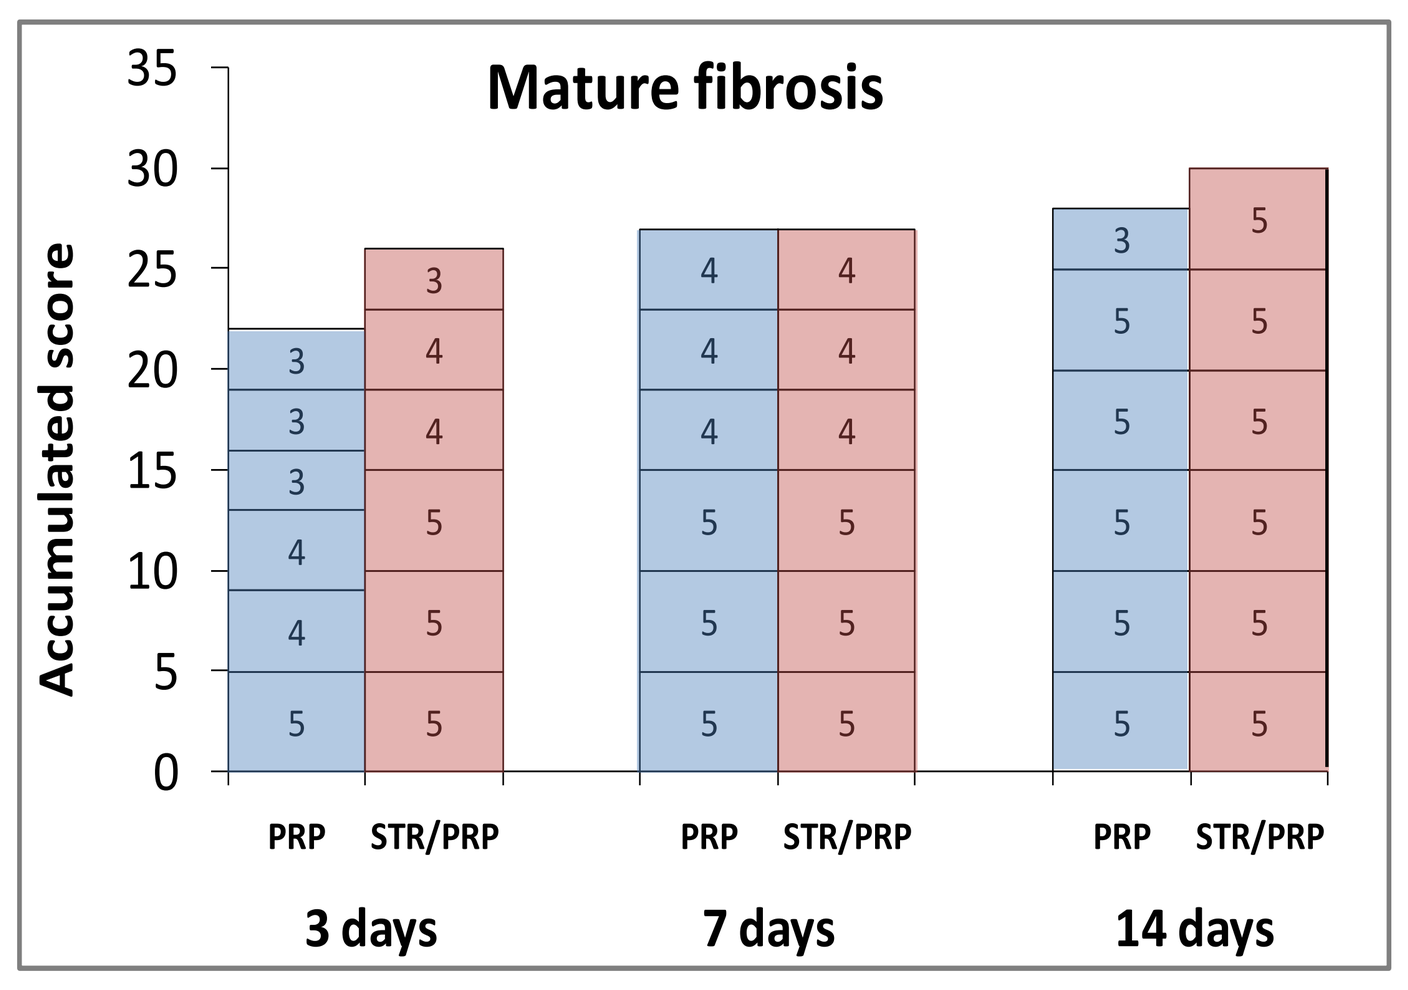

Mature fibrosis
Accumulated score
PRP STR/PRP
3 days
7 days
14 days

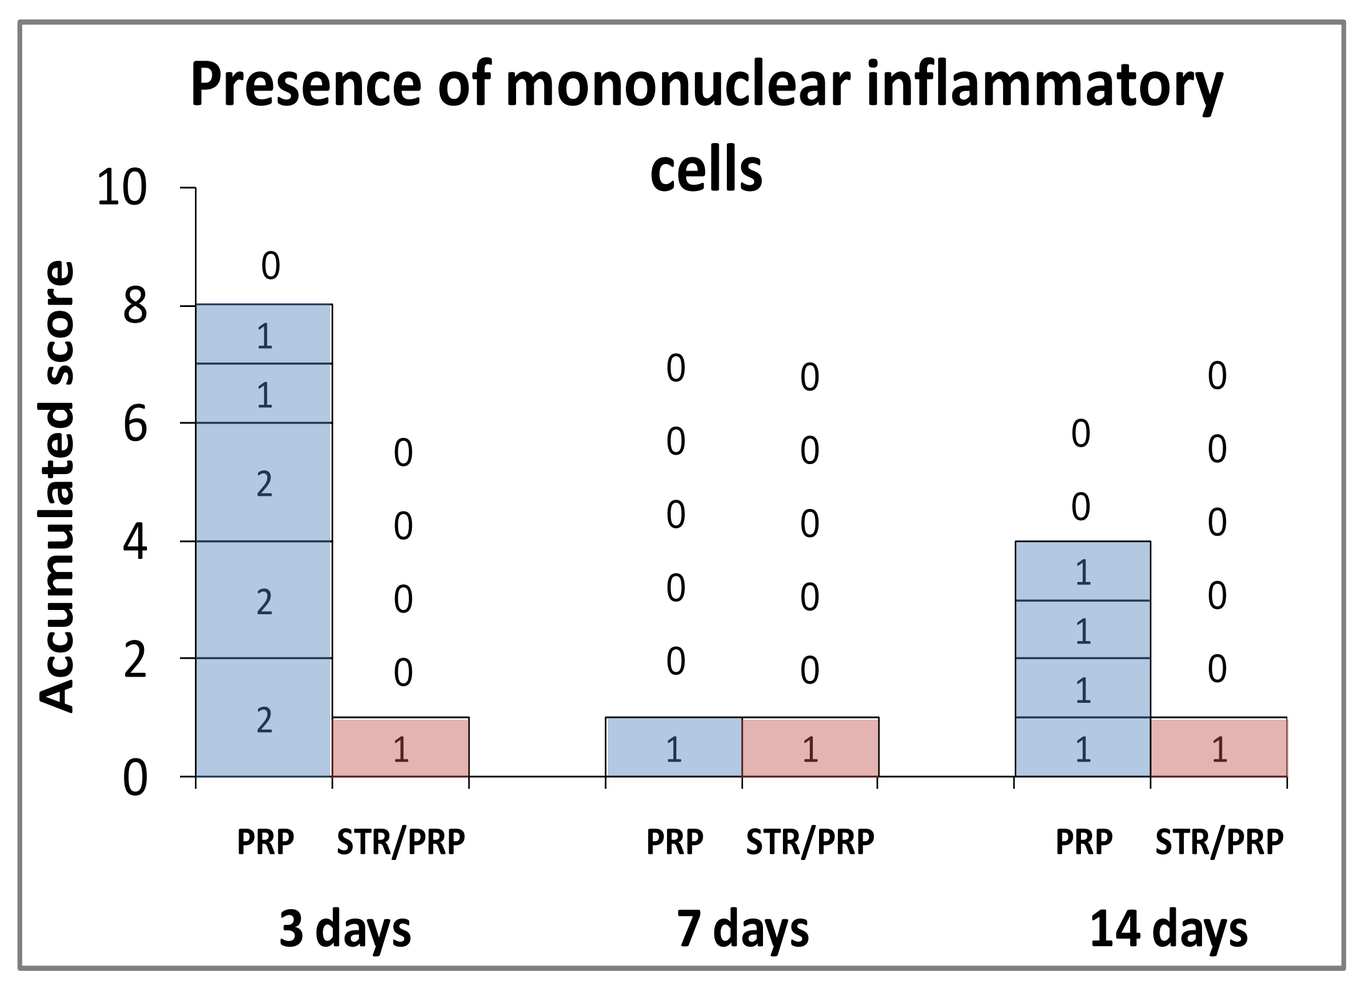

Presence of mononuclear inflammatory cells
Accumulated score
PRP STR/PRP
3 days
7 days
14 days

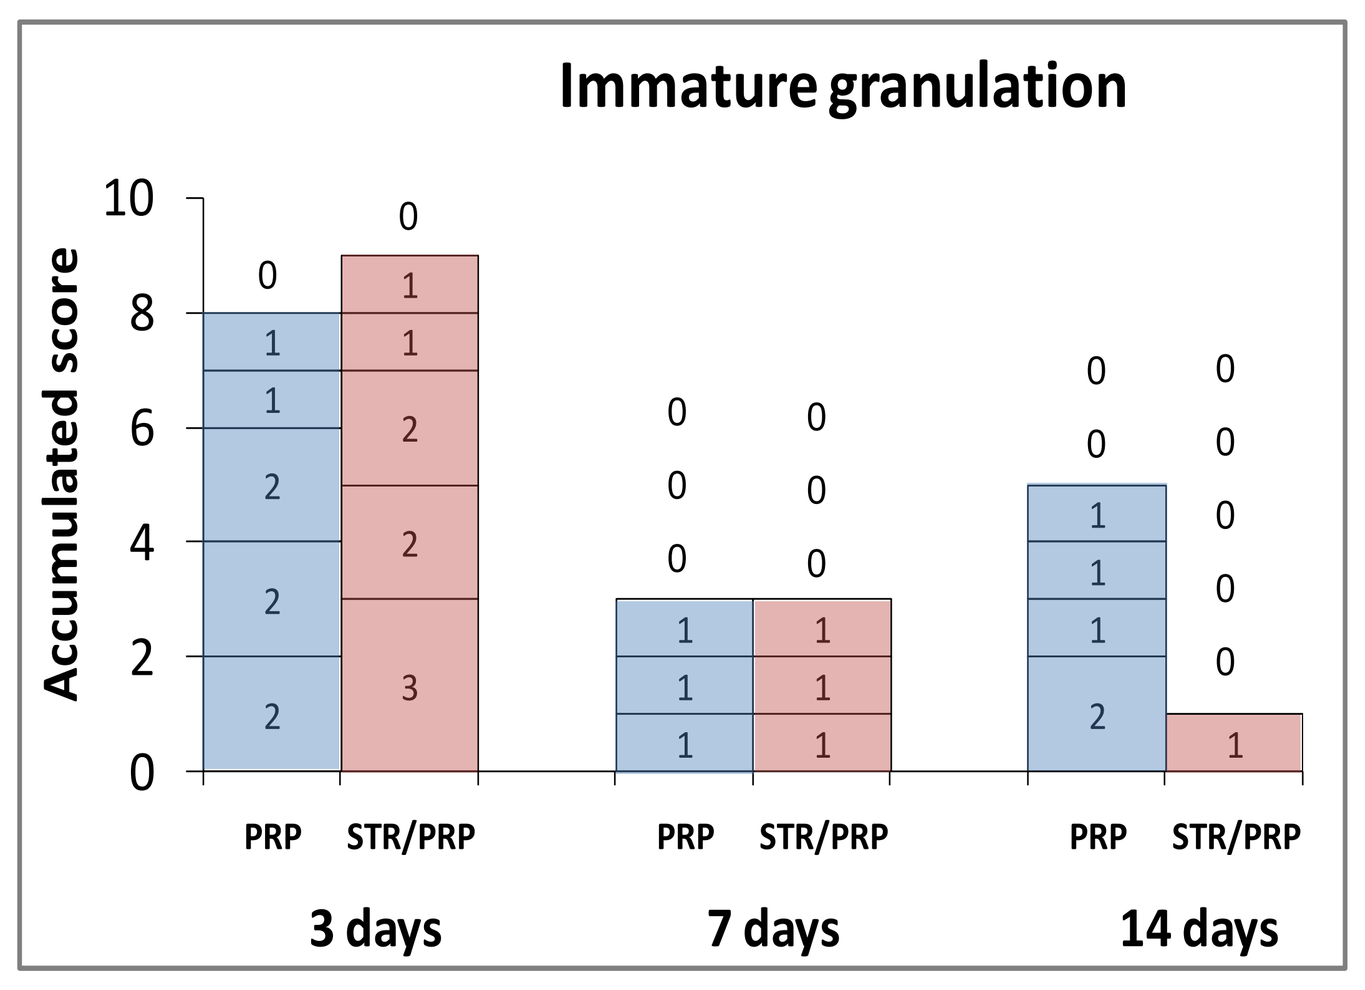

Immature granulation
Accumulated score
PRP STR/PRP
3 days
7 days
14 days

Summary and Conclusions

- ✓ Vergenix™STR is composed of human recombinant type I collagen expressed in transgenic tobacco plants
- ✓ Upon injection, Vergenix™STR combined with PRP forms *in situ* a matrix composed of collagen and fibrin
- ✓ STR/PRP matrix reduces inflammation and promotes cell proliferation and tissue healing
- ✓ STR/PRP matrix promotes *in situ* GFs generation and release while gradually degrading
- ✓ Both *in vivo* and *in vitro* studies showed superior performance of STR/PRP compared to PRP alone